Scan QRCode
to receive the poster!

Vector of innovation.

Customized methodology to investigate immuno-oncology drugs using humanized mouse models

Jean-François Mirjolet| Josselin Caradec| Olivier Duchamp| Francis Bichat| Damien France | Caroline Mignard | Fabrice Viviani
Oncodesign (France)

#1068

1 CONTEXT & OBJECTIVES

• Immunotherapy is one of the most exciting recent breakthroughs in the field of cancer treatment. Different approaches are developed such as cancer vaccines, adoptive cellular immunotherapy or immune checkpoint blockade, and a number have been regulatory approved or are currently investigated in clinic.

• Effective immunity against cancer involves complex interaction between the tumor, the host and the environment. Investigation and assessment of novel cancer immunotherapy approaches require the use of appropriate preclinical animal models that sufficiently reflect the physiological situation in cancer patients and that must be chosen carefully to address a specific scientific question.

• Human immune system is reconstituted in immunodeficient mice using either human PBMCs or hematopoietic stem cells (HSCs) as well as specific human immune cells such as Dendritic Cells (DCs), T cells, subset of T cells (e.g. gamma9 delta2 T cells) and NK cells. We also developed mouse humanization models using combinations of immune subpopulations such as co-transfer of autologous T cells and DCs.

• Humanized immune system (HIS) models were then used for in vivo evaluation of immunomodulatory agents with mice xenografted with cancer cell lines or patient-derived tumors. Therapeutic efficiency was assessed by following mice survival and tumor growth. The impact of therapeutics on tumors and immune cells was also assessed by flow cytometry and immunohistochemistry analyses.

• HIS models for research on cancer immunotherapy comprise three elements to define a custom-made preclinical model:

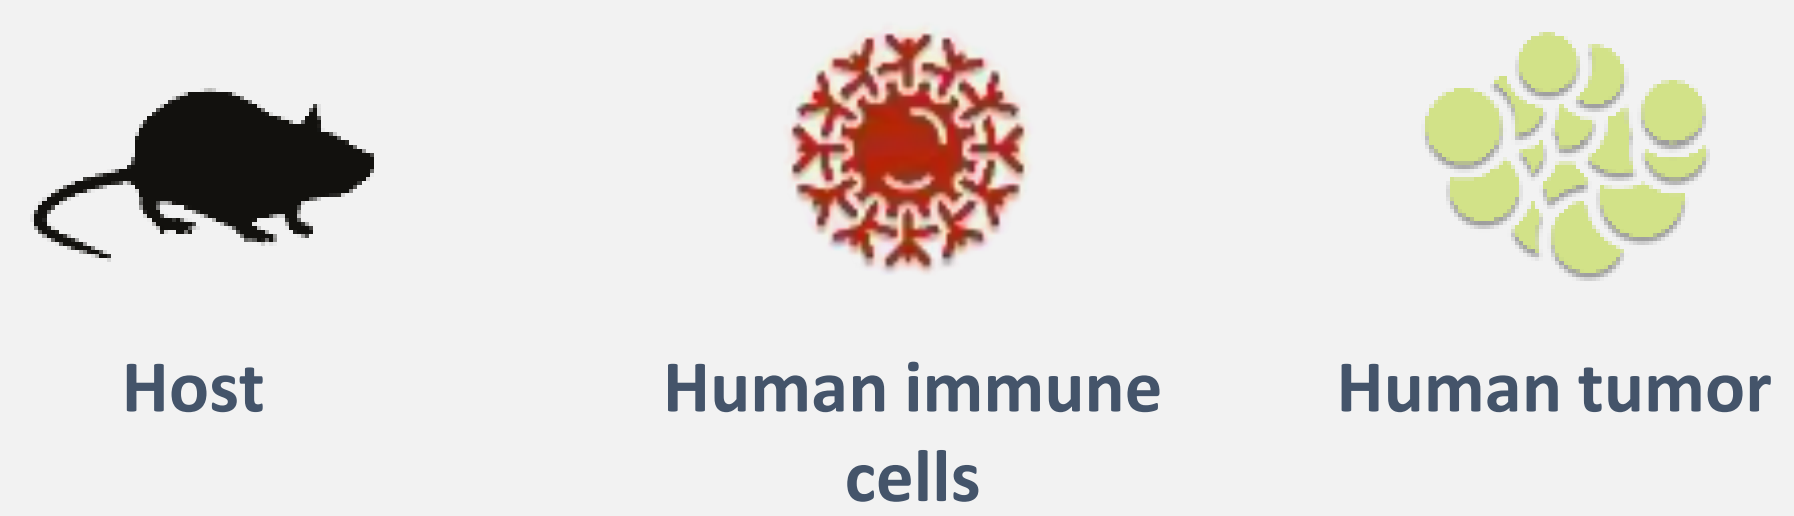
Host
Human immune cells
Human tumor

• Selection of each element to define the appropriate HIS model :

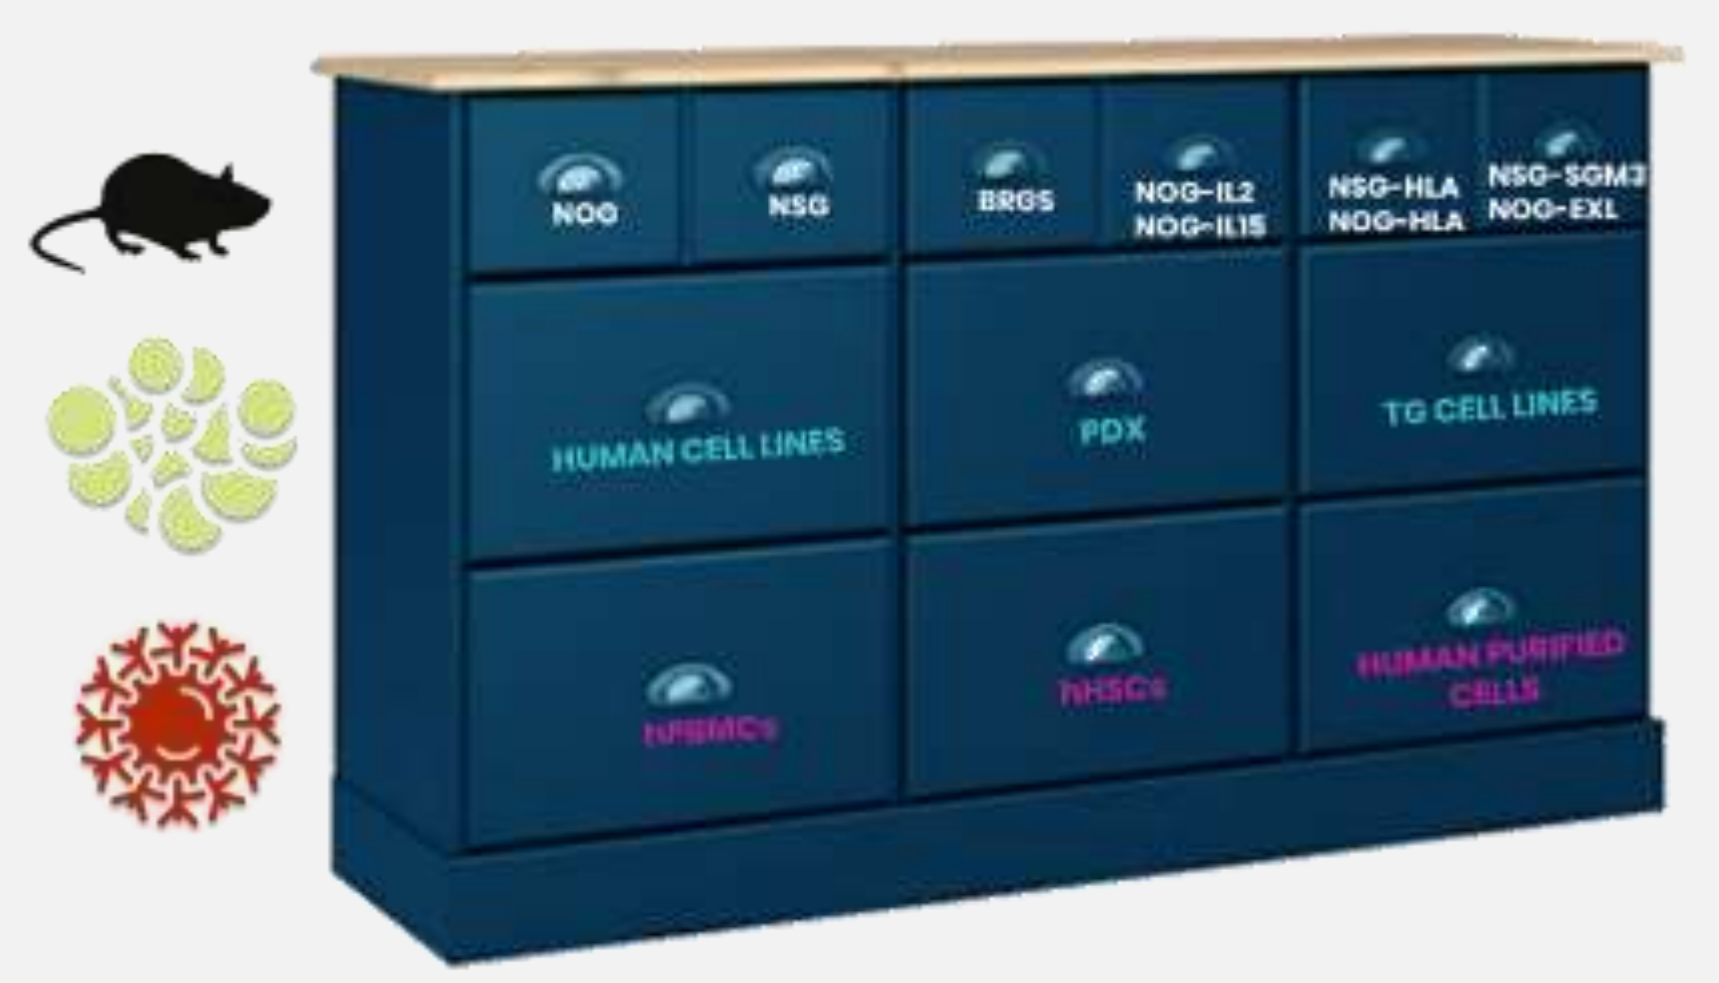
NOG
NSG
BRGS
NOG-IL2
NOG-IL15
NSG-HLA
NOG-HLA
NSG-SGM3
NOG-EXL
HUMAN CELL LINES
PDX
TG CELL LINES
hPBMCs
hHSCs
HUMAN PURIFIED CELLS

Parameters to consider for HIS model selection:
- MOA of test molecule
- Mouse host
- Tumor molecular and growth characteristics
- Type of human cells engrafted and supported
- Advantages/limitations of each HIS model

PBMC-engrafted mice
- Short-term studies
- Therapies targeting or mediated by T or NK cells
- Most appropriate for T cell bispecific
- Suitable for immune checkpoint inhibitors

HSC-engrafted mice (NOG, NSG, BRG..)
- Long-term studies
- Therapies targeting or mediated by T cells
- Suitable for immune checkpoint inhibitors

HSC-engrafted mice (NOG-EXL, NSG-SGM3, BRGS...)
- Long-term studies
- Therapies targeting or mediated by myeloid-lineage cells
- Therapies targeting or mediated by T cells
- Suitable for immune checkpoint inhibitors

2 RESULTS

In vivo evaluation of engineered T cells

Hematologic tumor (IV)

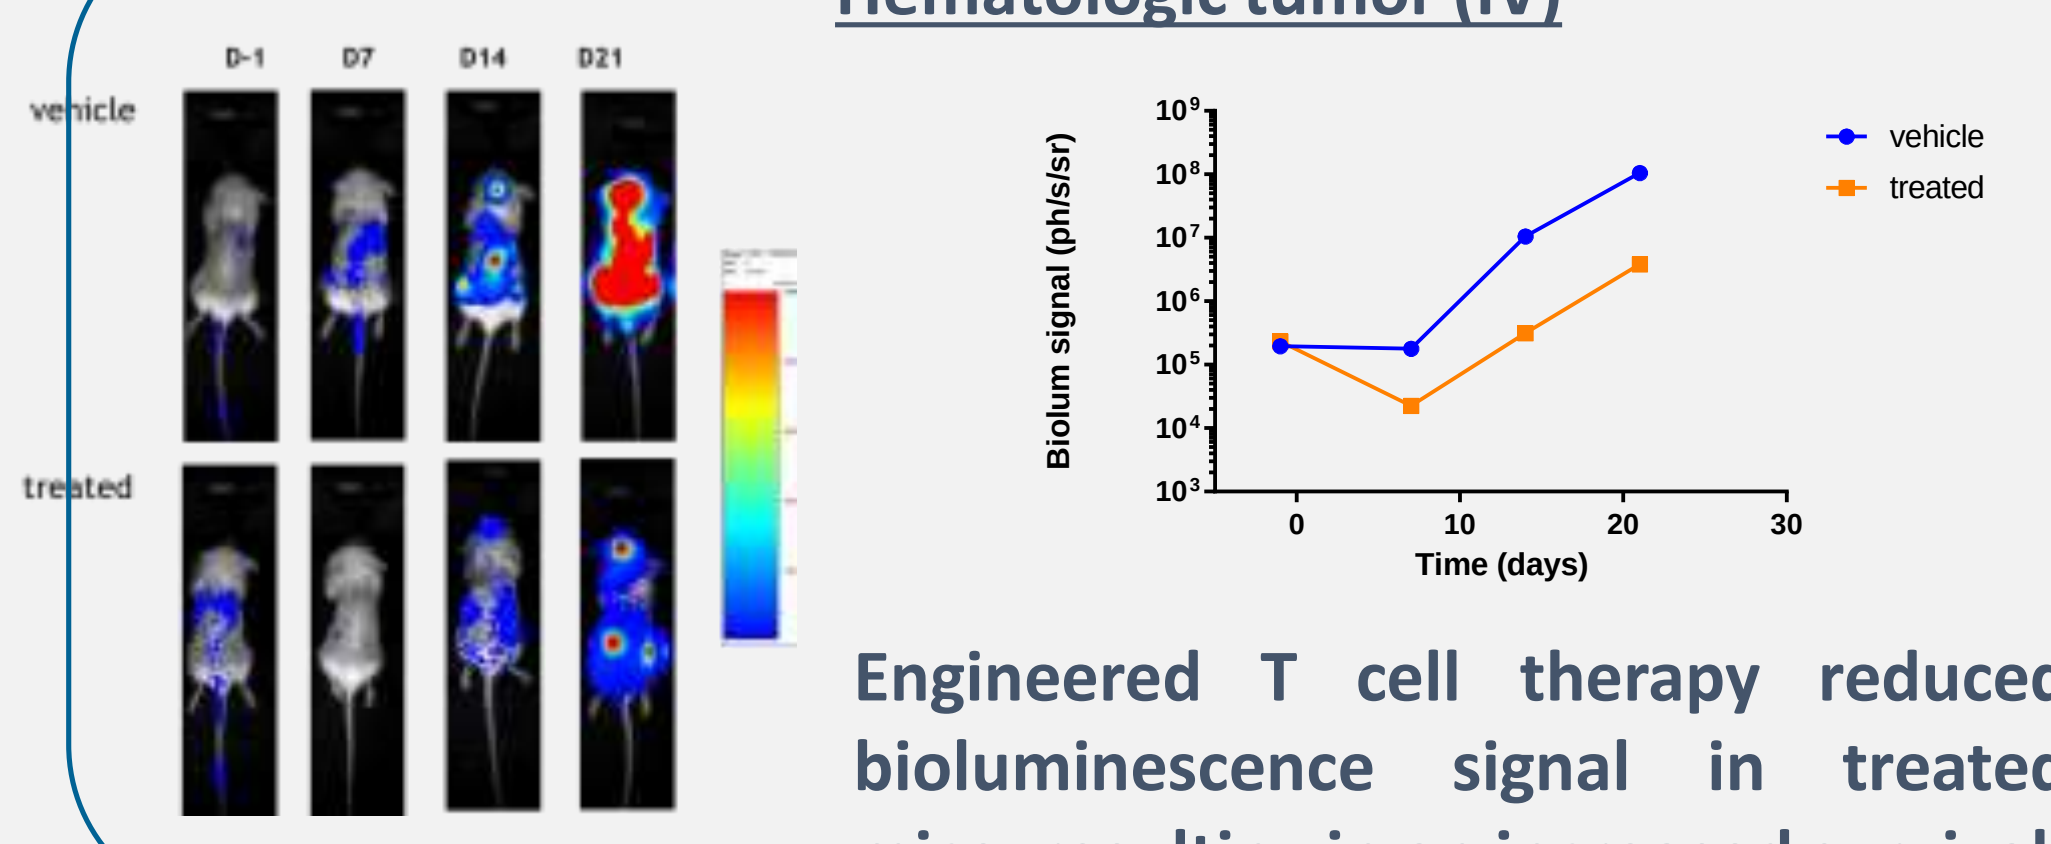
D-1
D7
D14
D21
vehicle
treated
Biolum signal (ph/s/sr)
Time (days)

Engineered T cell therapy reduced bioluminescence signal in treated mice, resulting in an increased survival

Solid tumor (SC)

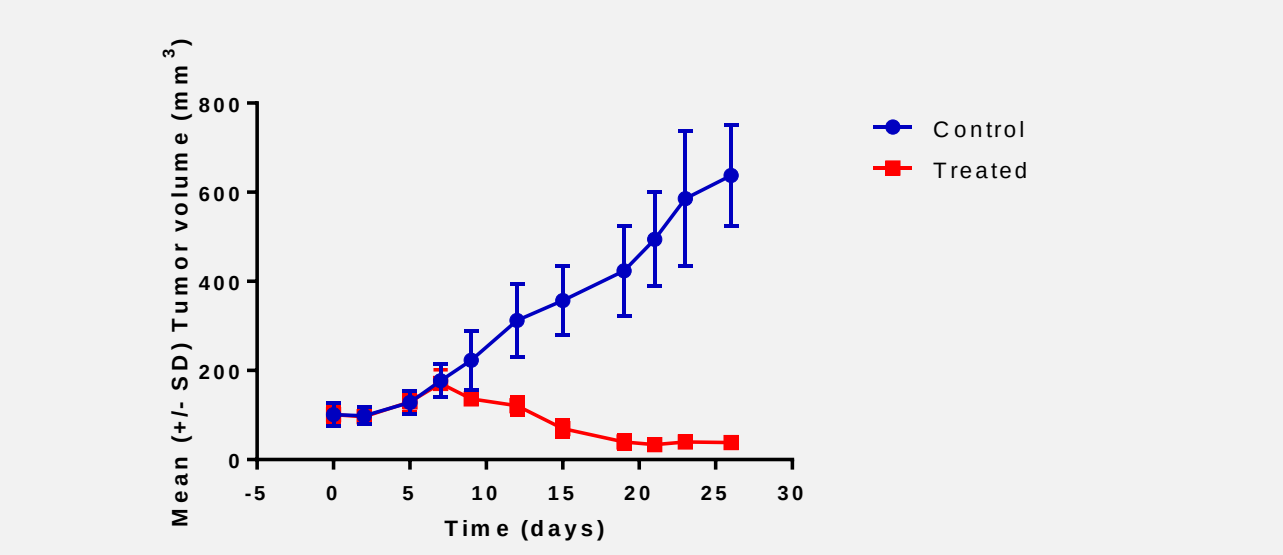
Mean (+/- SD) Tumor volume (mm3)
Control
Treated
Time (days)

Engineered T cell therapy induced tumor regression

In vivo evaluation of T cell engager antibodies

Circulating biomarker

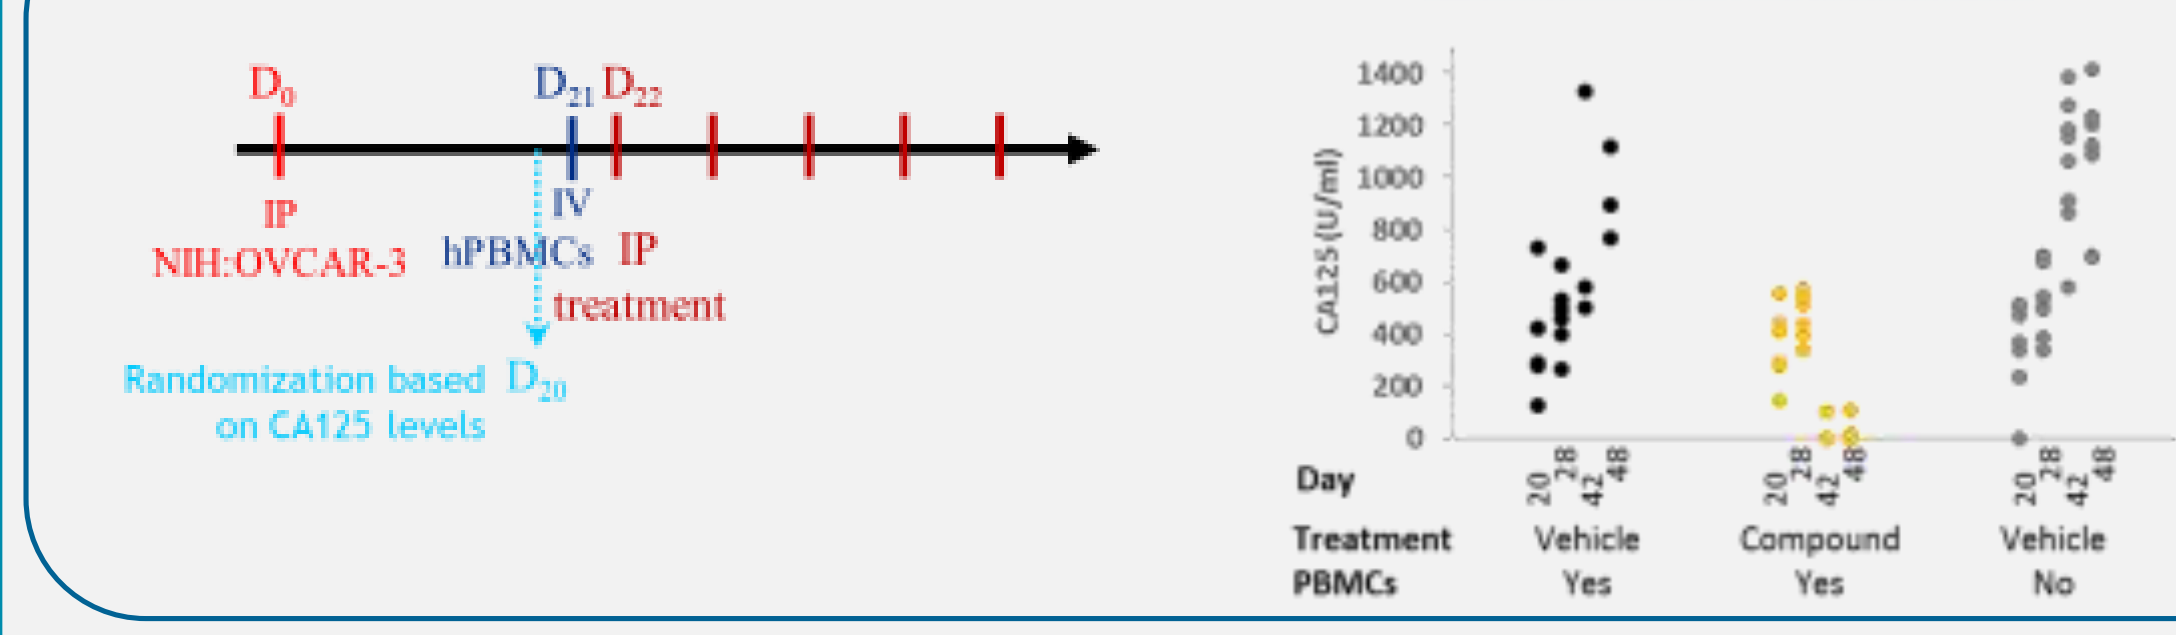
D0
D21 D22
IP
NIH:OVCAR-3
IV
hPBMCs
IP
treatment
Randomization based on CA125 levels
D20
CA125 (U/ml)
Day
Treatment
PBMCs
Vehicle
Yes
Compound
Yes
Vehicle
No

Decrease of CA125 circulating biomarker in ovarian tumor correlates with a significant antitumor activity of bispecific T-cell engager antibody.

Tumor volume

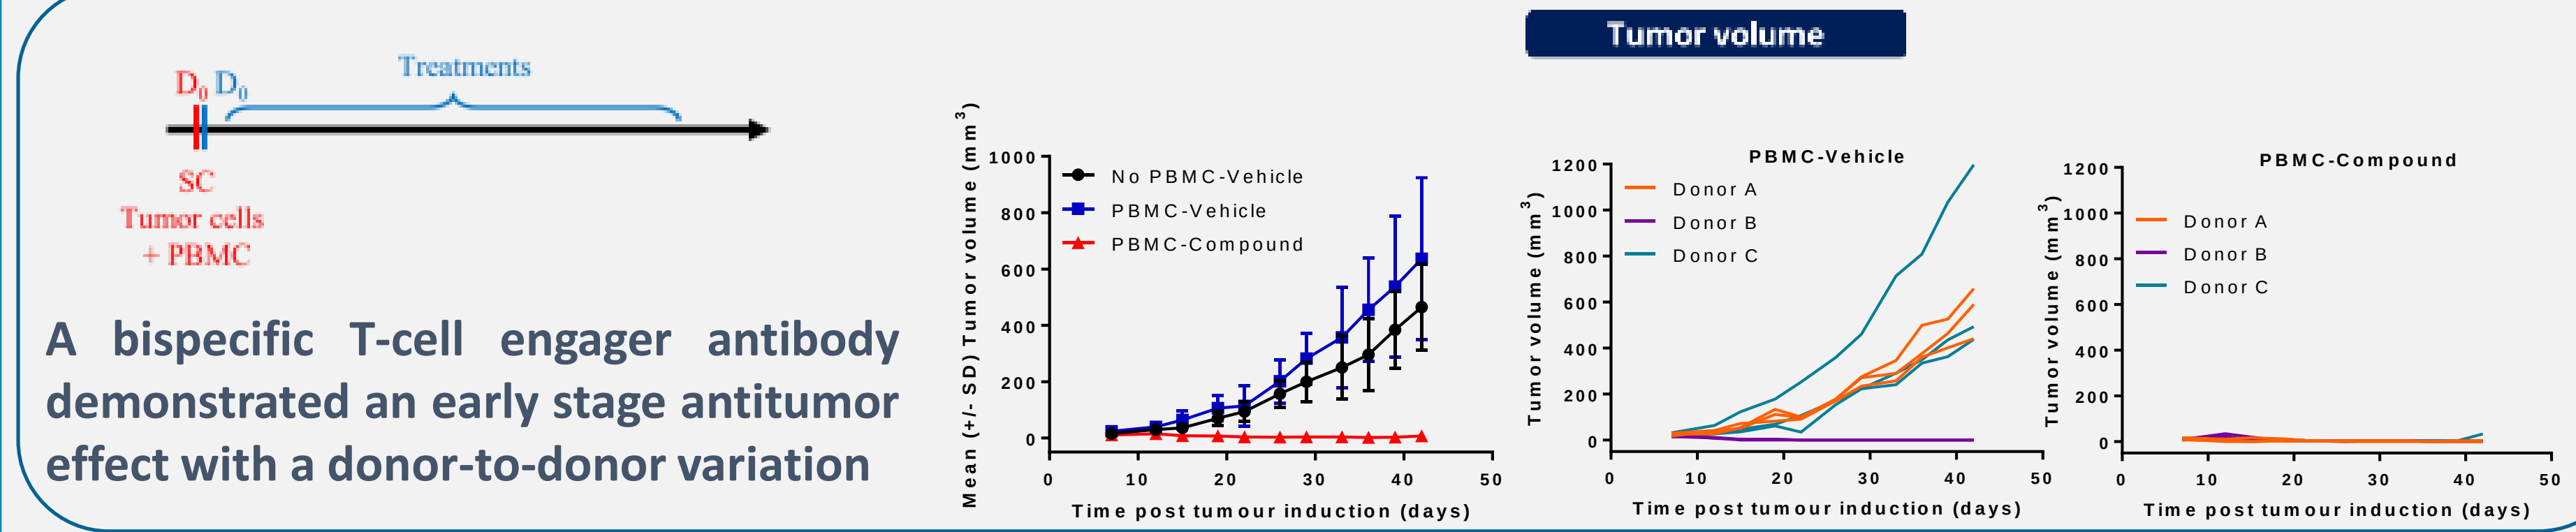
D0 D0
Treatments
SC
Tumor cells
+ PBMC
No PBMC-Vehicle
PBMC-Vehicle
PBMC-Compound
Mean (+/- SD) Tumor volume (mm3)
Time post tumour induction (days)
PBMC-Vehicle
PBMC-Compound
Donor A
Donor B
Donor C
Tumor volume (mm3)

A bispecific T-cell engager antibody demonstrated an early stage antitumor effect with a donor-to-donor variation

Heme tumor / **Solid tumor**

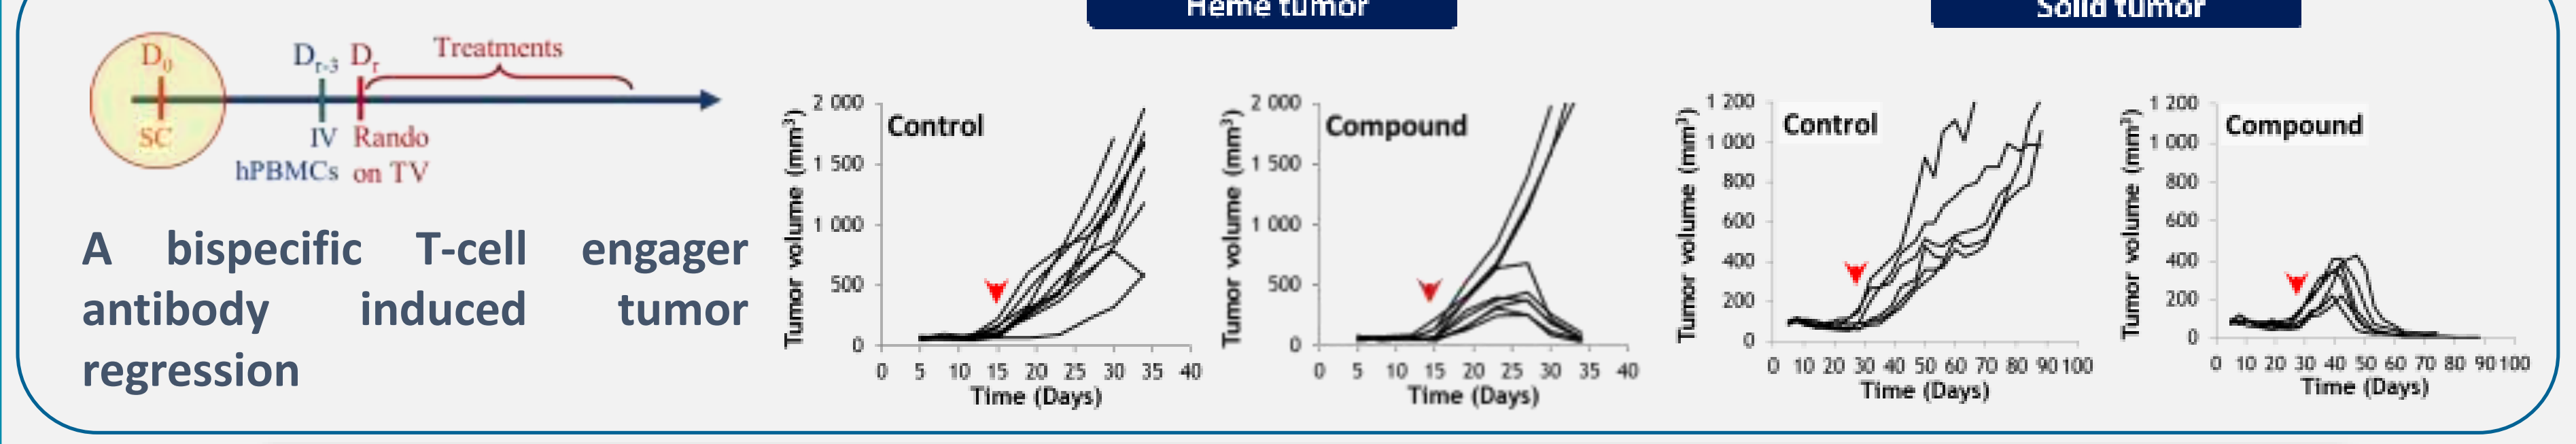
D0
SC
Dr-3 Dr
IV
hPBMCs
Rando on TV
Treatments
Control
Compound
Tumor volume (mm3)
Time (Days)

A bispecific T-cell engager antibody induced tumor regression

In vivo evaluation of compound targeting antigen presenting cells

Autologous T cells and moDCs + tumor cell line

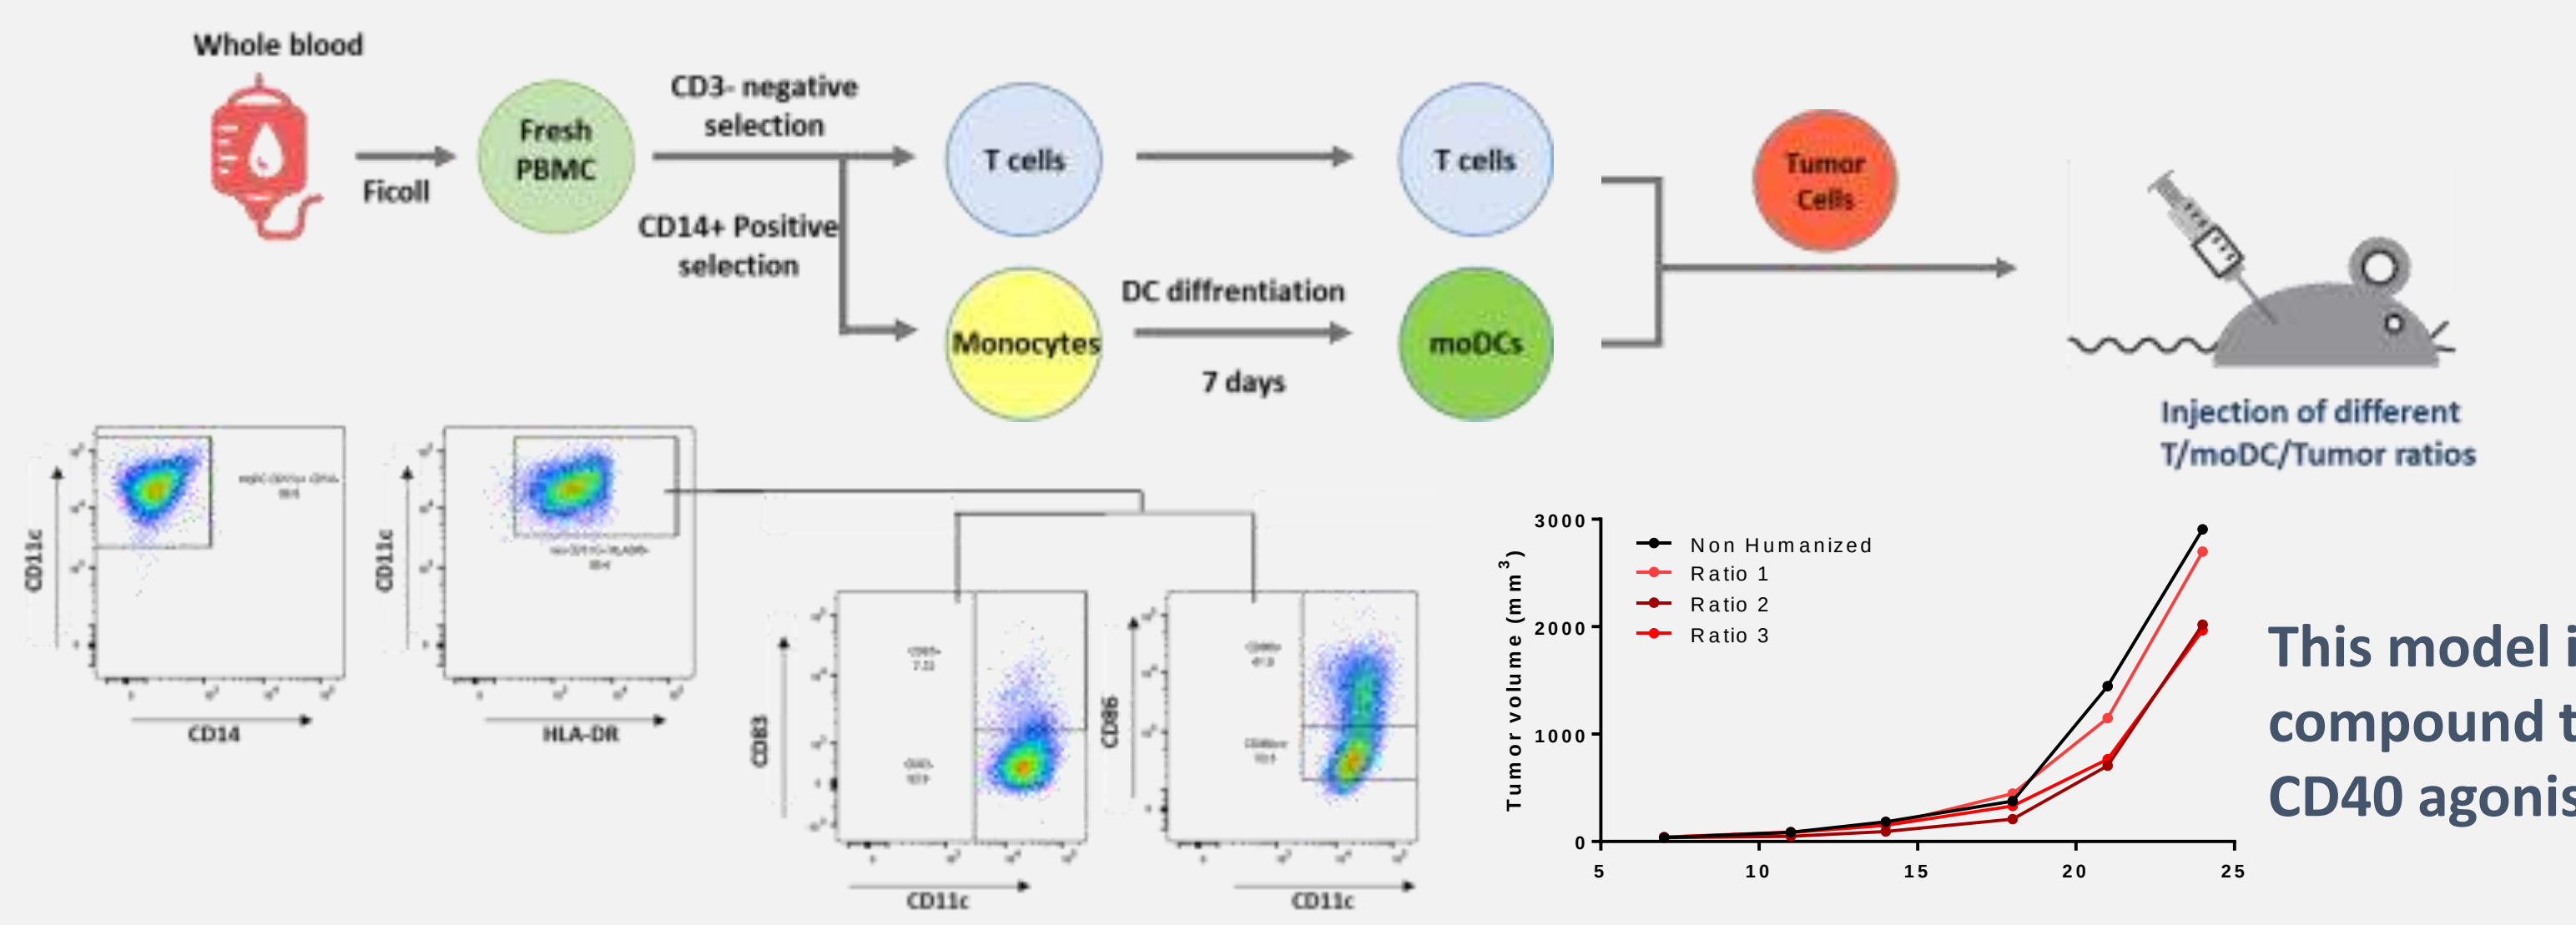
Whole blood
Ficoll
Fresh PBMC
CD3- negative selection
CD14+ Positive selection
T cells
T cells
Monocytes
DC diffrentiation
7 days
moDCs
Tumor Cells
Injection of different T/moDC/Tumor ratios
CD11c
CD14
HLA-DR
CD83
CD86
Non Humanized
Ratio 1
Ratio 2
Ratio 3
Tumor volume (mm3)

This model is useful to evaluate compound targeting DC such as CD40 agonist

3 RESULTS

In vivo evaluation of immune checkpoint modulators

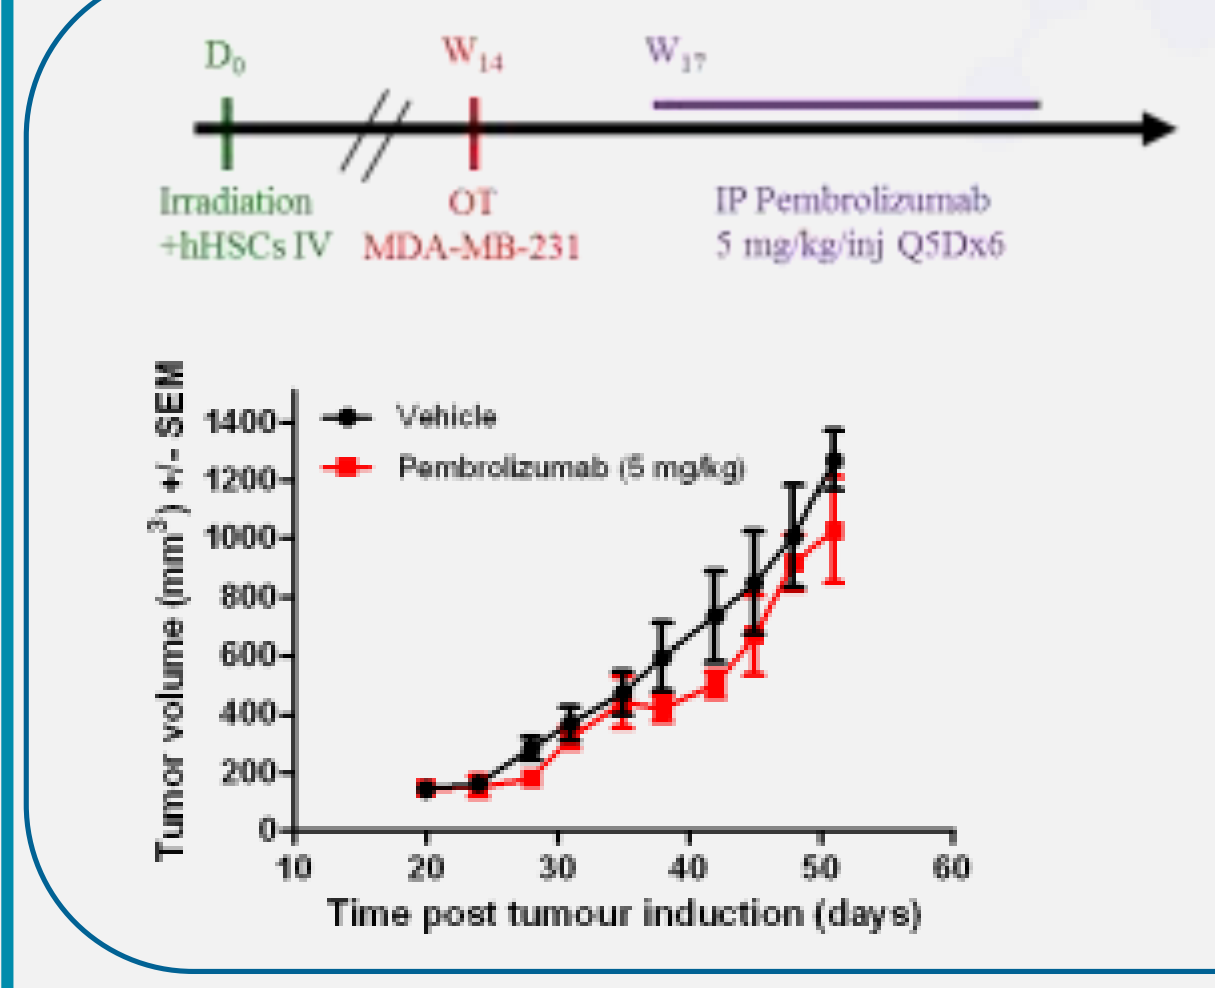
D0
W14
W17
Irradiation +hHSCs IV
OT MDA-MB-231
IP Pembrolizumab 5 mg/kg/inj Q5Dx6
Tumor volume (mm3) +/- SEM
Vehicle
Pembrolizumab (5 mg/kg)
Time post tumour induction (days)

Tumor growth is reduced in Pembrolizumab-treated mice with a maximum TGI of 30%. An increased proportion of T cells was evidenced inside the tumor from Pembrolizumab- treated mice

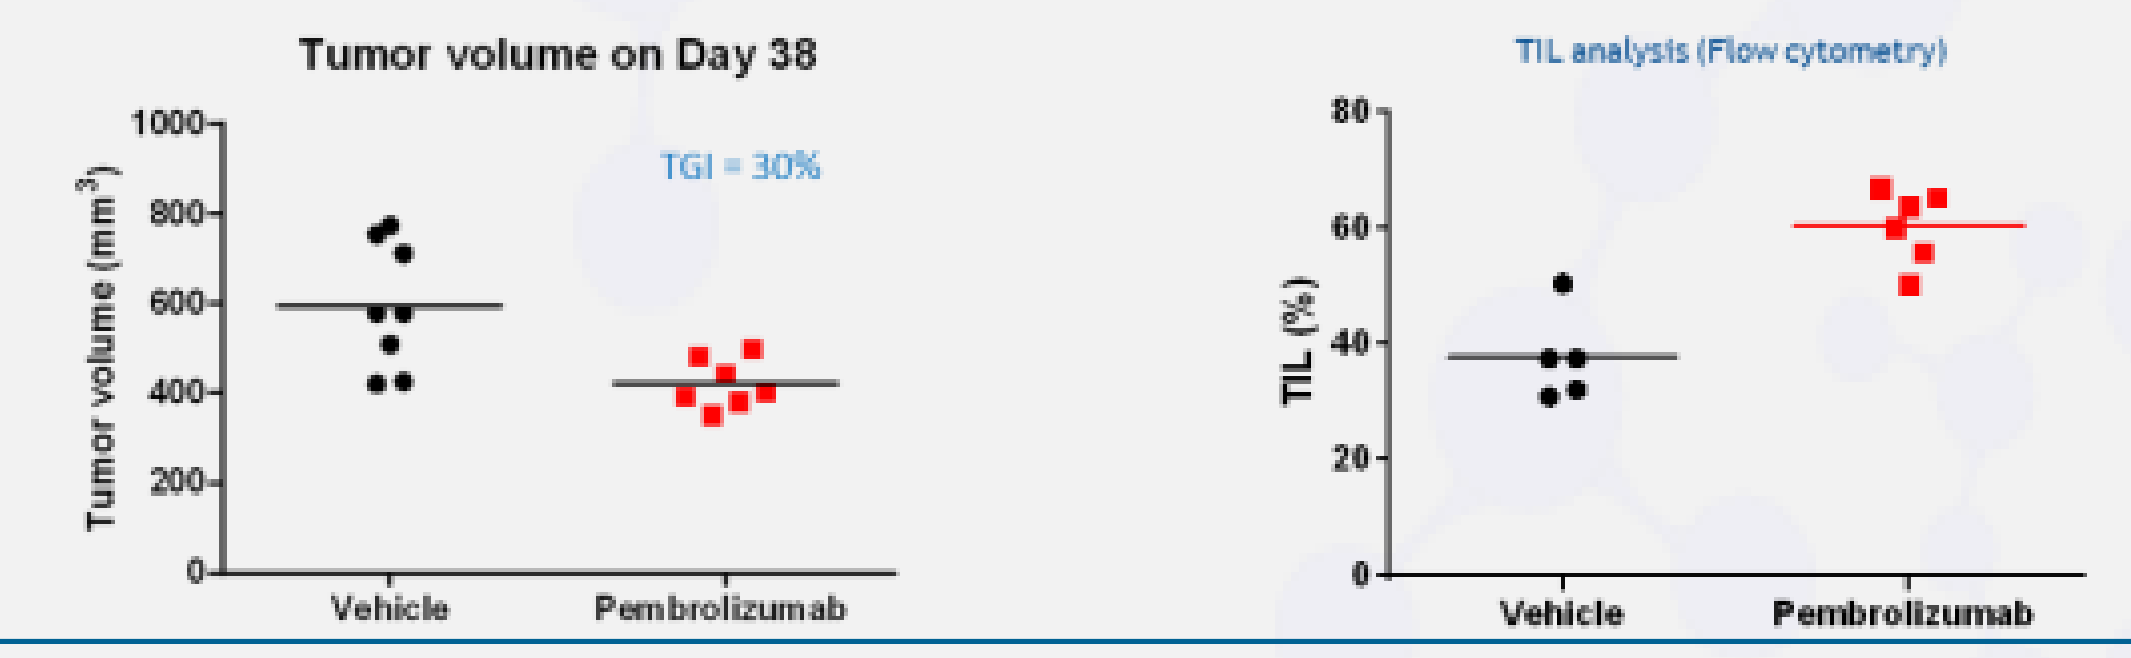
Tumor volume on Day 38
TGI = 30%
Tumor volume (mm3)
Vehicle
Pembrolizumab
TIL analysis (Flow cytometry)
TIL (%)

ID# PDX	Patient Infiltrat	NOG-PBMC	NOG-EXL (HSCs)
OD-BRE-589	+/-	-	-
IM-BRE-044	+++	++	+++
IM-OVA-512	+/-	+/-	+/-
IM-OVA-535	+++	+/-	+

ID# PDX	Mouse model	hCD45	hCD3	hCD8	hFoxP3	hPD-L1	CD163
OD-BRE-589 (TNBC, poorly infiltrated in patient)	NOG-PBMC	-	-	-	-	-	ND
	NOG-EXL (HSCs)	-	-	-	-	-	ND
IM-BRE-044 (TNBC, highly infiltrated in patient)	NOG-PBMC	++	++	++	+	++	-
	NOG-EXL (HSCs)	+++	+++	+++	+++	++	+++

IM-BRE-044 /NOG-EXL (HSC) mice

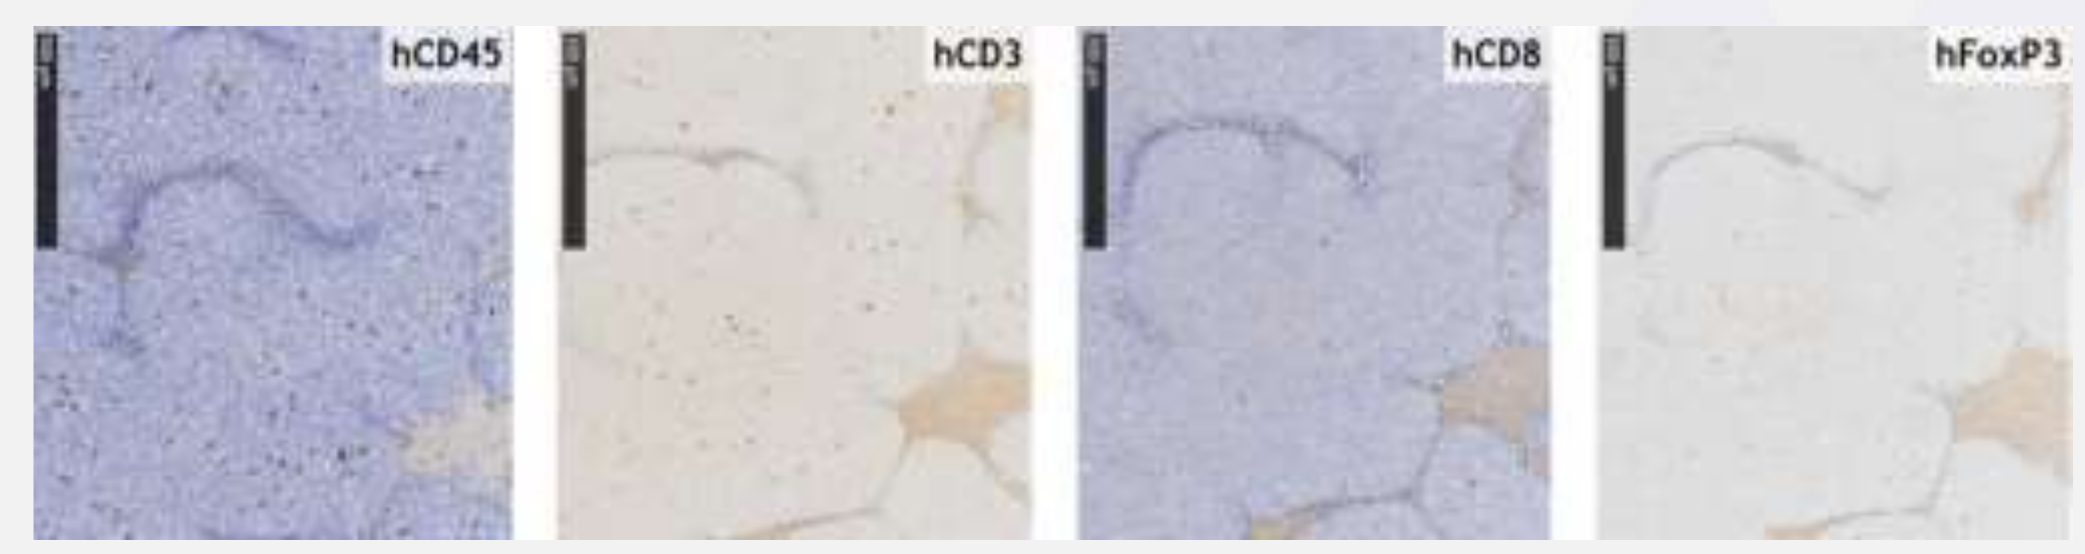
hCD45
hCD3
hCD8
hFoxP3

• Infiltration in mice is correlated with immune cell infiltration in originating patient tumor.

• Pembro therapy did not induced tumor volume reduction under these experimental conditions. Additional work is on-going.

Tumor volume

IM-BRE-044/NOG-PBMC mice — IM-BRE-044 /NOG-EXL (HSC) mice

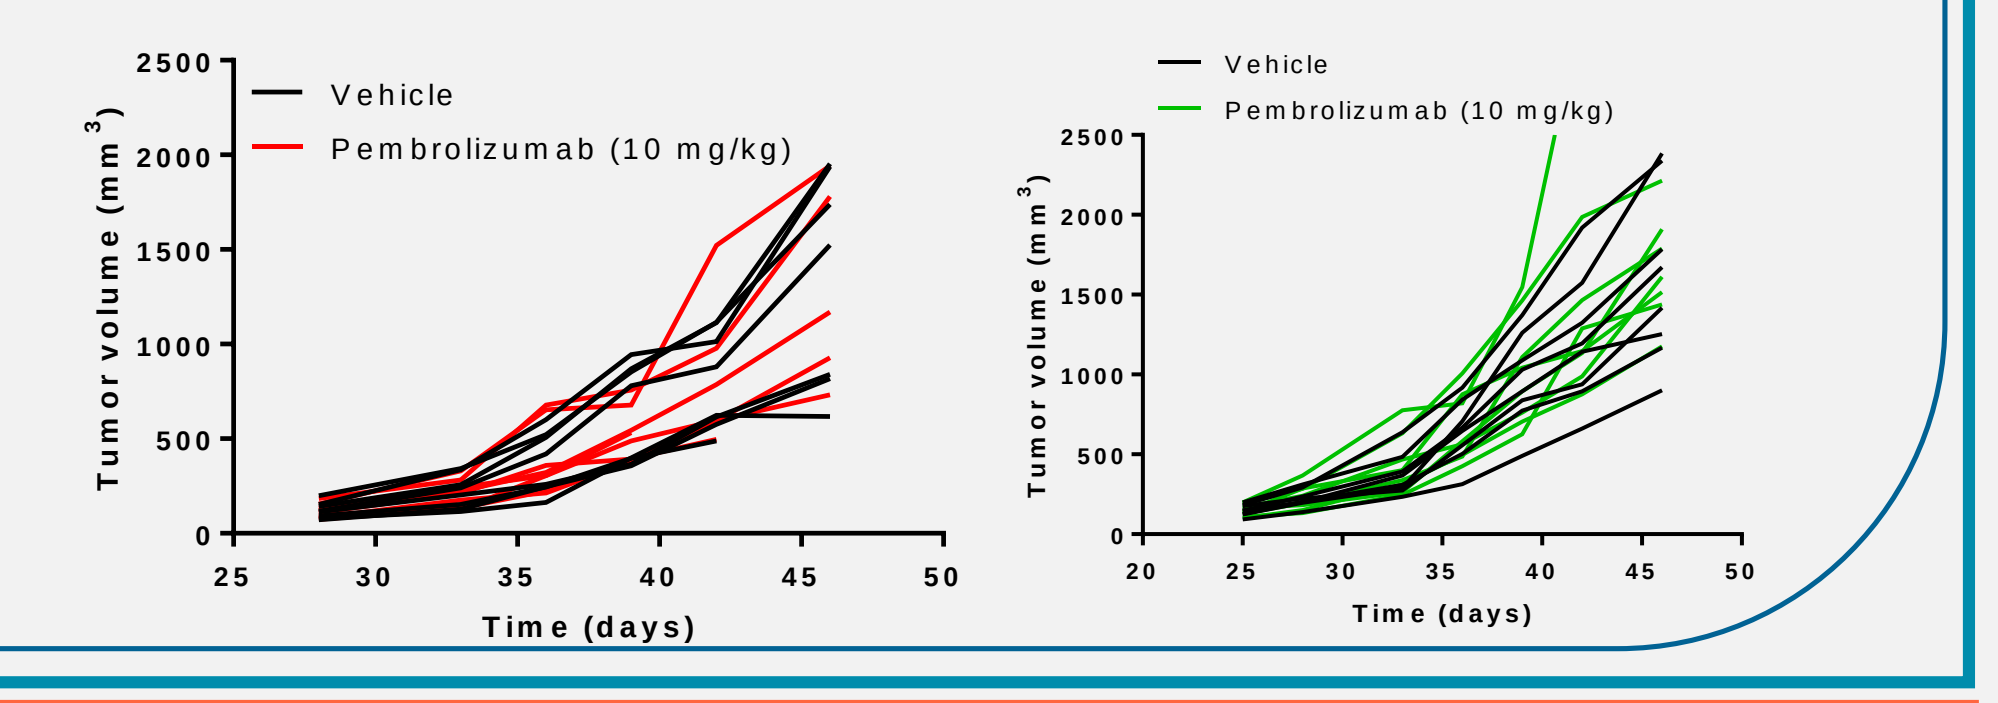
Vehicle
Pembrolizumab (10 mg/kg)
Tumor volume (mm3)
Time (days)

4 CONCLUSION

• There is no unique HIS tumor model that mirrors the tumor in patient and the complexity of the cancer microenvironment, implying the most appropriate model to be considered and selected to answer a specific question.
• PDX models harboring both a human immune system and a tumor fragment from the same patient may lead us towards personalized medicine and may be useful for drug development in immuno-oncology. These models are currently in development.

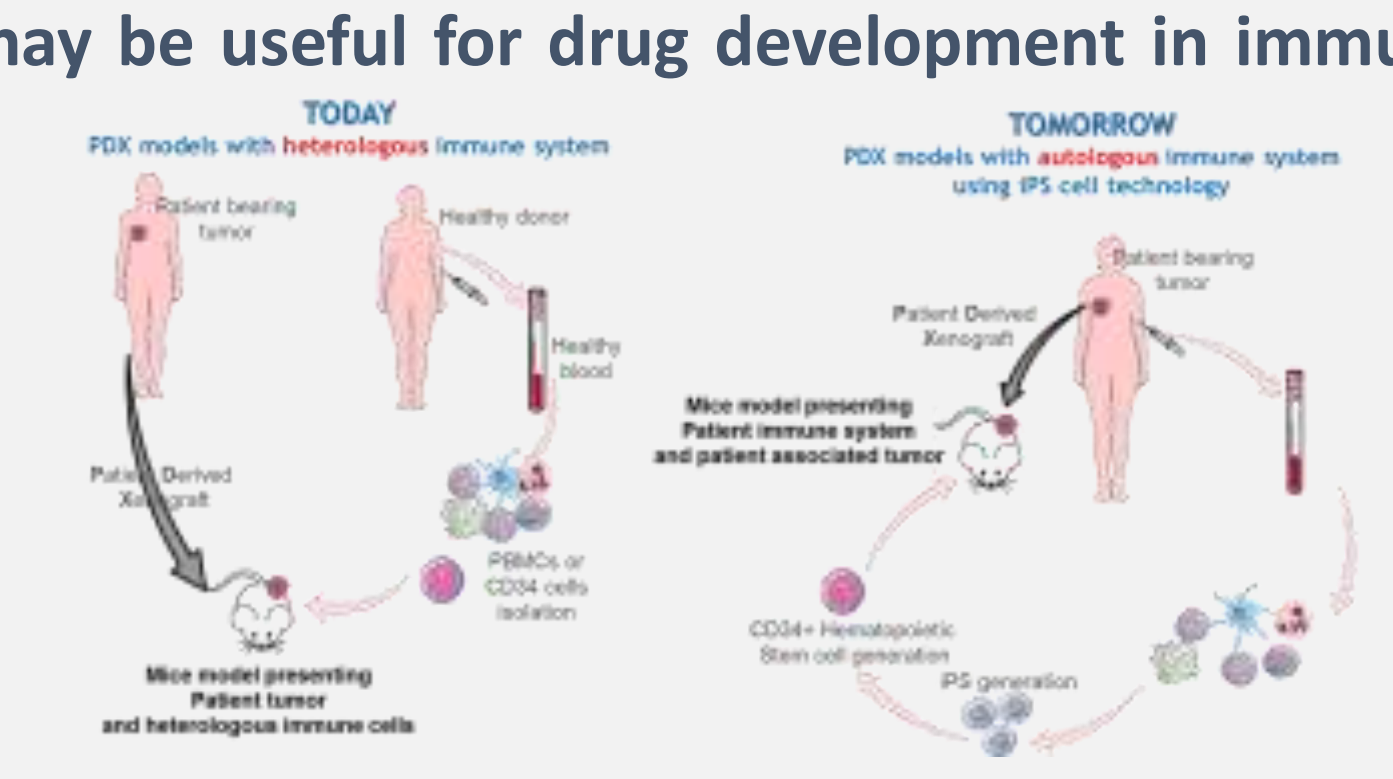
TODAY
PDX models with heterologous immune system
TOMORROW
PDX models with autologous immune system using iPS cell technology

AACR |Atlanta | April 2019

oncodesign.com